Poster #114

Treatment-Emergent Adverse Events (TEAEs) in Children With Ataxia-Telangiectasia Treated for One Year With Intra-Erythrocyte Dexamethasone Sodium Phosphate (EryDex)

Biljana Horn, Dirk Thye, Maureen Roden
Quince Therapeutics, South San Francisco, CA

INTRODUCTION

- Ataxia-telangiectasia (A-T) is an inherited rare neurodegenerative and immunodeficiency disorder caused by pathogenic variants of the *ATM* gene
- Previous short-term, small studies suggest that treatment with corticosteroids may lead to neurological improvements in patients with A-T[1-5]
- Due to short half-lives, standard corticosteroids require frequent dosing to provide efficacy
- High peak drug levels from intermittent dosing are related to significant long-term side effects such as hyperglycemia, immunosuppression, and suppression of the hypothalamic-pituitary-adrenal (HPA) axis
- EryDex is a novel investigational agent designed to provide continuous delivery of steroids to patients who require prolonged use
- EryDex releases dexamethasone gradually from erythrocytes, which is believed to reduce toxicity by eliminating peak levels

OBJECTIVE

Evaluate treatment-emergent adverse events (TEAEs) in patients with A-T treated with EryDex for 1 year (NCT02770807) compared to placebo control.

METHODS

EryDex System

- Autologous Intracellular Drug Encapsulation (AIDE) technology allows dexamethasone sodium phosphate to be encapsulated in autologous erythrocytes
- Components of the EryDex System:
 1. Red Cell Loader **(Figure 1)**:
 Non-invasive active medical device that automates the EryDex System (EDS) process and handles blood, drug, and processing solutions
 2. EryKit: Disposable, single-use, sterile kit includes bags and lines to contain blood, drug, and processing solutions

Figure 1. Red Cell Loader

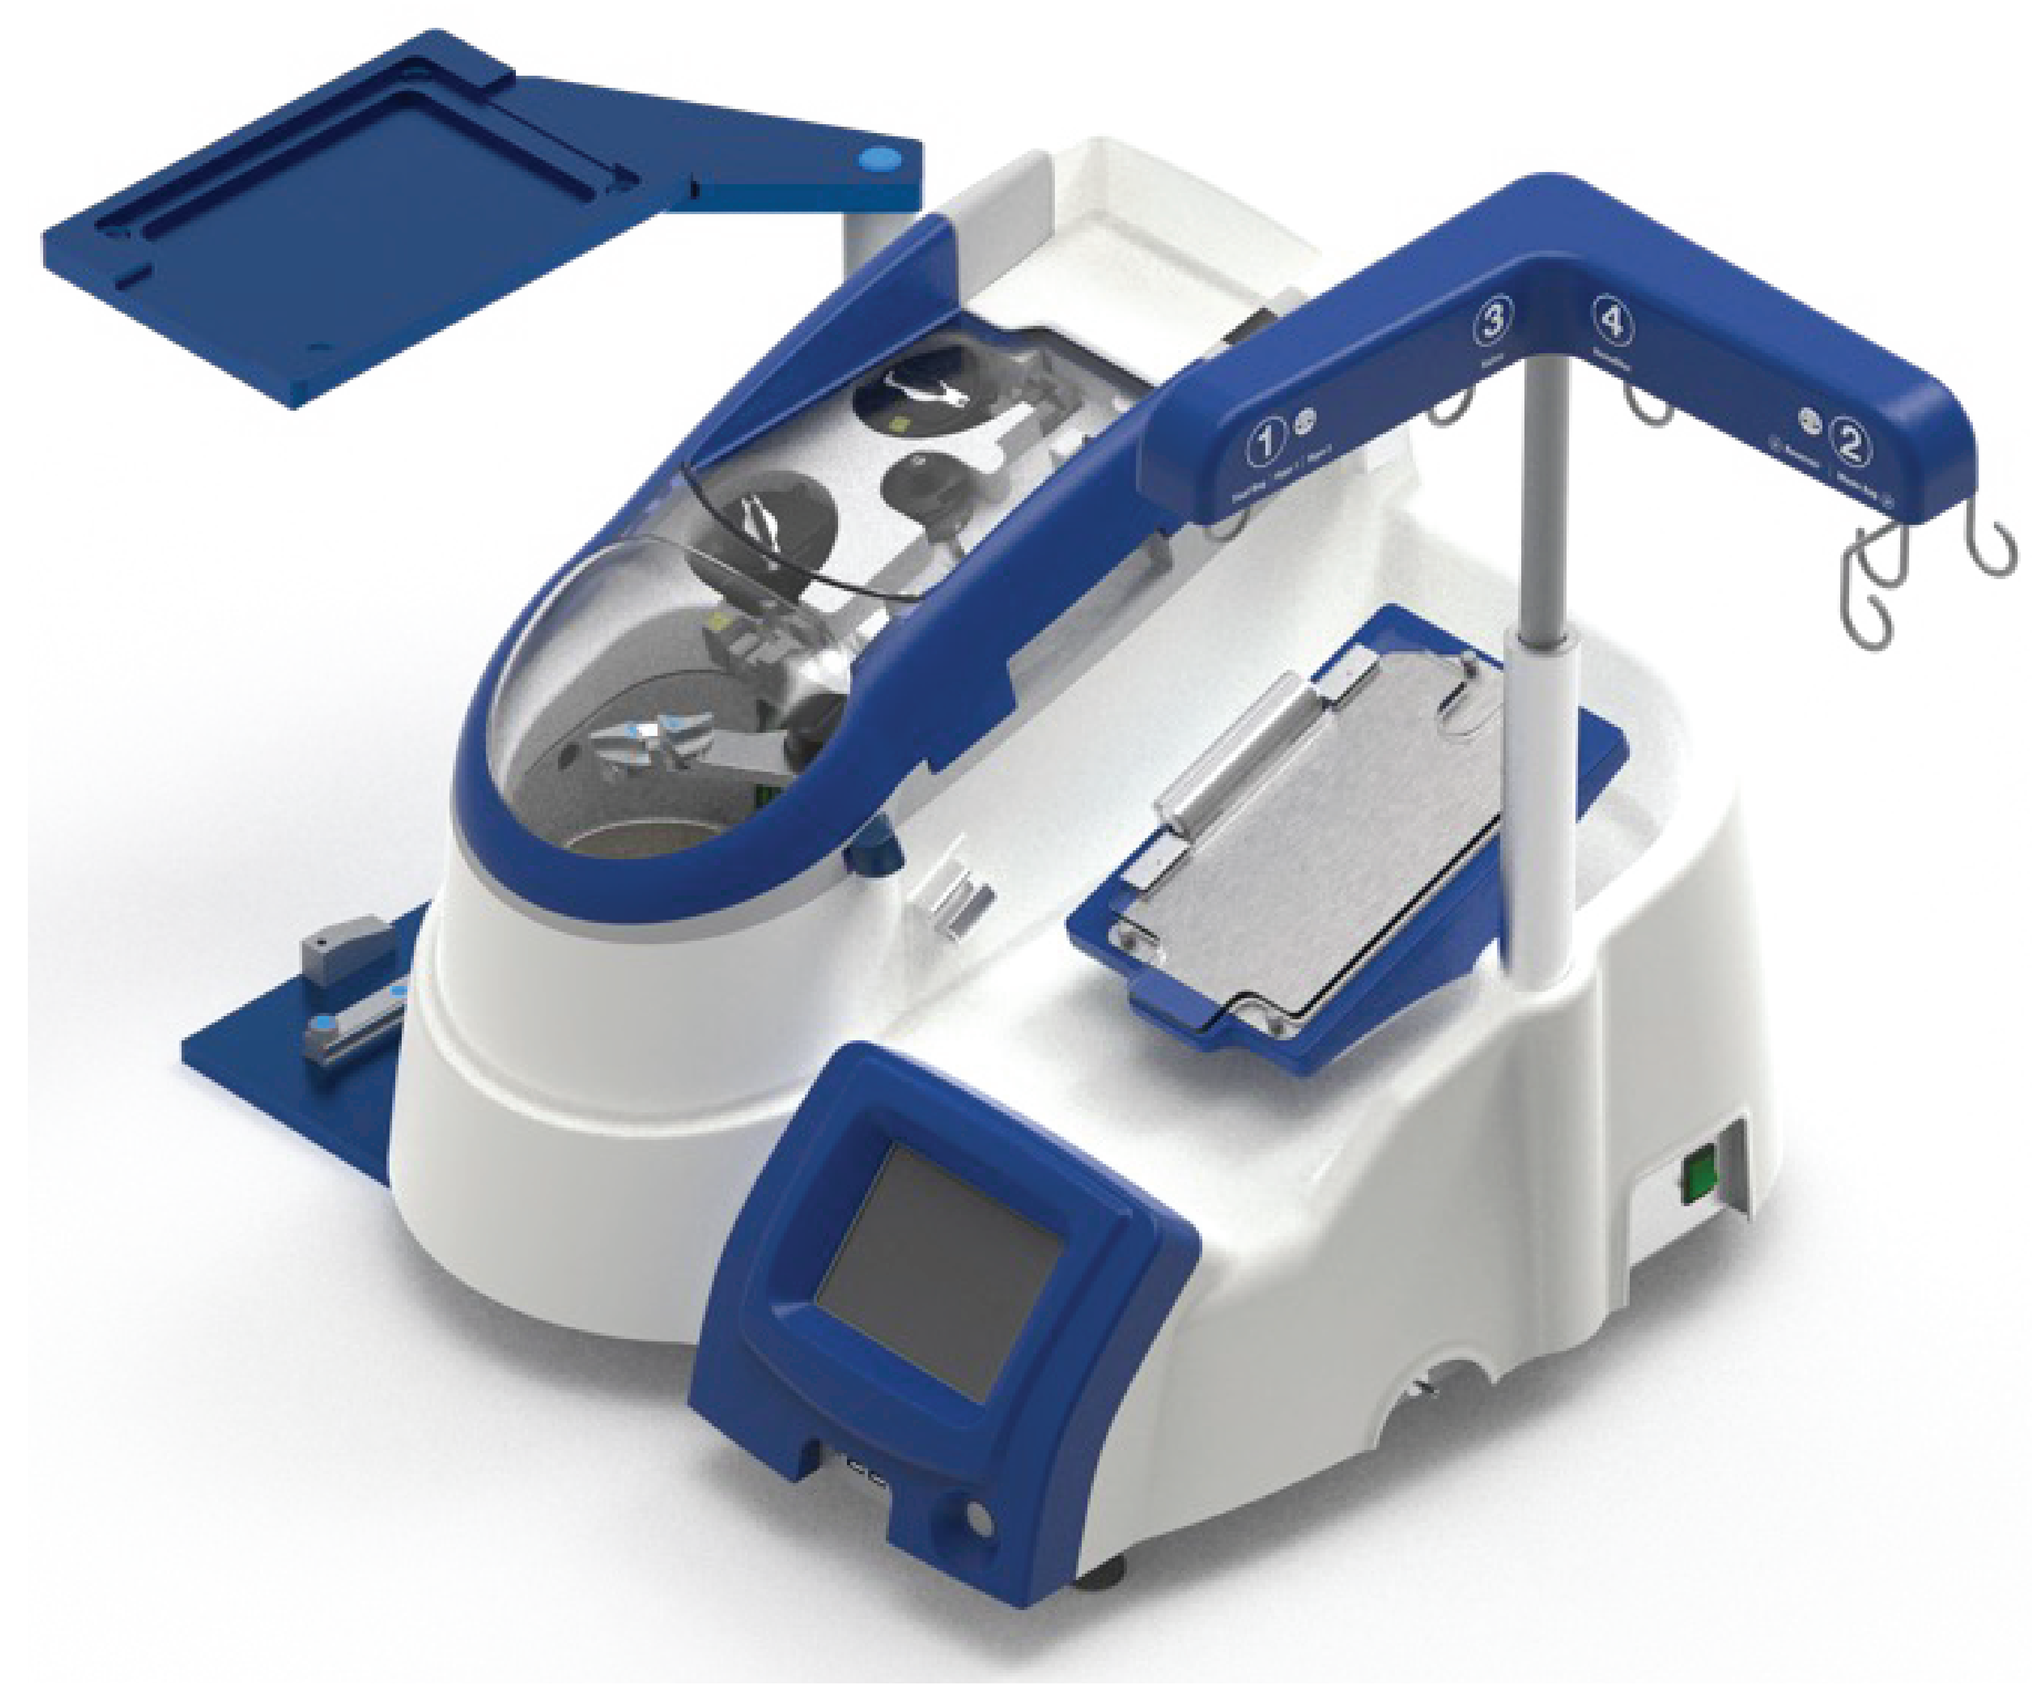

EryDex System Process

- Designed to be administered monthly at a hospital or treatment center

Phase 1: Blood Collection
- 50 mL of patient blood is collected

Phase 2: Blood Processing
- Red Cell Loader is set up by a trained operator with EryKit and Process Solutions

Phase 3: EryDex Infusion
- EryDex (drug-loaded red blood cells [RBCs]) is immediately (within 30 minutes) infused using a standard blood infusion set

References: 1. Buoni S, Zannolli R, Sorrentino L, Fois A. Betamethasone and improvement of neurological symptoms in ataxia-telangiectasia. *Arch Neurol.* 2006;63(10):1479-1482. **2.** Broccoletti T, Del Giudice E, Amorosi S, et al. Steroid-induced improvement of neurological signs in ataxia-telangiectasia patients. *Eur J Neurol.* 2008;15(3):223-228. **3.** Broccoletti T, Del Giudice E, Cirillo E, et al. Efficacy of very-low-dose betamethasone on neurological symptoms in ataxia-telangiectasia. *Eur J Neurol.* 2011;18(4):564-570. **4.** Zannolli R, Buoni S, Betti G, et al. A randomized trial of oral betamethasone to reduce ataxia symptoms in ataxia telangiectasia. *Mov Disord.* 2012;27(10):1312-1316. **5.** Hasegawa S, Kumada S, Tanuma N, et al. Long-term evaluation of low-dose betamethasone for ataxia telangiectasia. *Pediatr Neurol.* 2019;100:60-66. **6.** Zielen S, Crawford T, Benatti L, et al. Safety and efficacy of intra-erythrocyte dexamethasone sodium phosphate in children with ataxia telangiectasia (ATTeST): a multicentre, randomised, double-blind, placebo-controlled phase 3 trial. *Lancet Neurol.* 2024;23(9):871-882.

STUDY DESIGN

- The ATTeST study (NCT02770807) is a phase 3, multicenter, randomized, double-blind, placebo-controlled trial evaluating the effects of intra-erythrocyte dexamethasone sodium phosphate on neurological symptoms in patients with A-T[6]
- 175 ambulatory patients with A-T (aged ≥6 years; weight >15 kg) randomized (1:1:1) to receive monthly IV infusion of either:
 - EryDex ~5-10 mg (low dose)
 - EryDex ~14-22 mg (high dose)
 - Placebo
 - At Months 6 and 9, one-third of patients on placebo switched to active drug
 - Treatment continued for 12 months
 - Adverse events (AEs) reported during 12 months of treatment were coded using MedDRA version 24.0

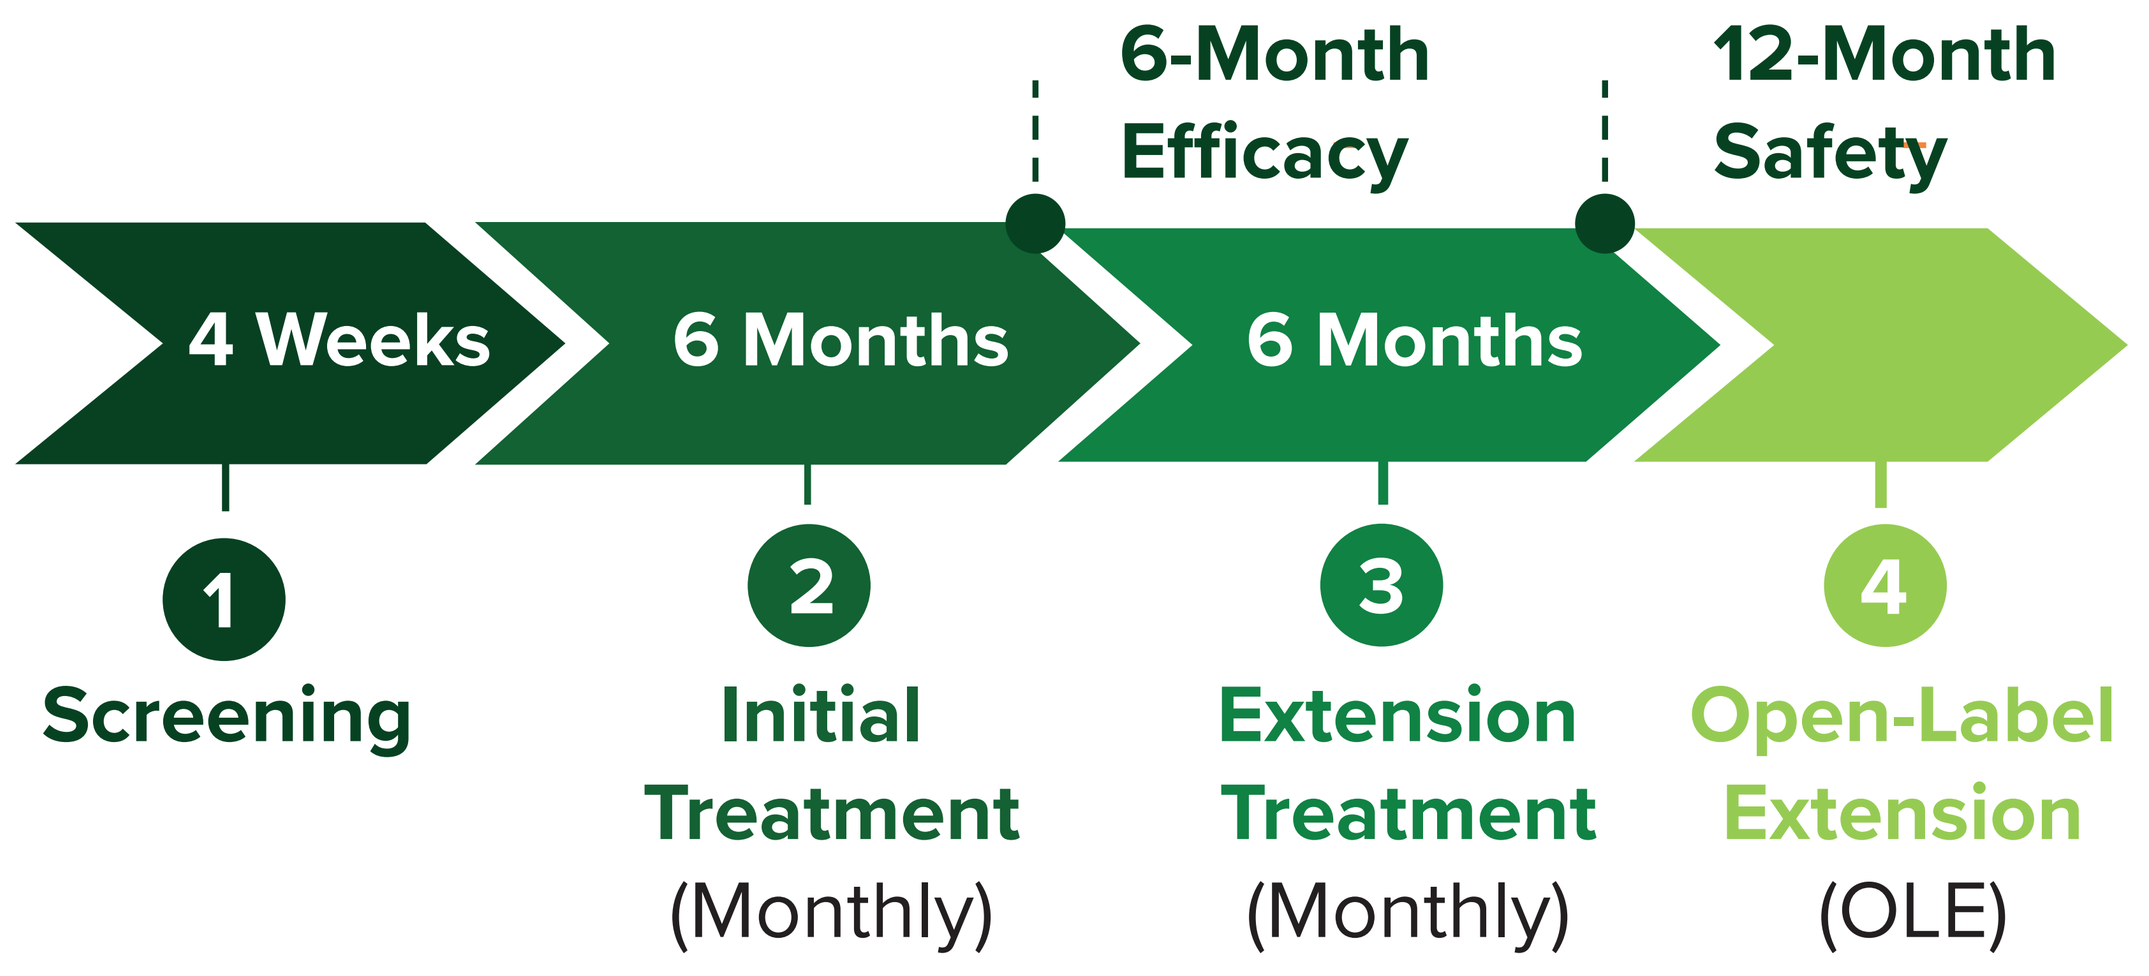

6-Month Efficacy
12-Month Safety
4 Weeks
6 Months
6 Months
1
Screening
2
Initial Treatment (Monthly)
3
Extension Treatment (Monthly)
4
Open-Label Extension (OLE)

RESULTS

Table 1. Reported Treatment-Emergent Adverse Events (TEAEs)

	Initial 6-Month Treatment Period			Through Month 12		
Patients with:	Placebo (N=59)	EryDex Low Dose (N=59)	EryDex High Dose (N=57)	Non-Switch Placebo (N=19)	EryDex Low Dose (N=59)	EryDex High Dose (N=57)
Any TEAE (%)	73%	73%	82%	79%	76%	88%
Any Treatment-Related TEAE (%)	25%	25%	37%	26%	32%	44%
Any Serious TEAE (%)	12%	10%	12%	21%	14%	16%
Any Serious Treatment-Related TEAE (%)	0	0	2%	5%	2%	2%
Any TEAE Leading to Discontinuation (%)	0	0	4%	0	2%	4%
Any TEAE Leading to Death (%)	0	0	0	0	0	0

- The proportion of patients who experienced at least 1 TEAE was comparable across the 3 arms through 12 months of treatment **(Table 1)**
- EryDex treatment was generally well tolerated, with most adverse events (AEs) being mild to moderate and transient
- Three patients were discontinued from the study: 1 patient in the low-dose group had a serious adverse event (SAE) of B-cell lymphoma (unlikely treatment-related) and 2 patients in the high-dose group had TEAEs of pyrexia and tachycardia (1 patient, probably treatment-related) and pain/pruritus (1 patient, possibly treatment-related)
- There were no TEAEs leading to death

Table 2. TEAEs Occurring in >10% of EryDex-Treated Patients

- Reported TEAEs occurring in >10% of patients were similar between EryDex- and placebo-treated patients with the exception of pruritus **(Table 2)**
- Pruritus was noted only in EryDex-treated patients; pruritus is a well-known complication occurring during intravenous infusion of dexamethasone phosphate
- Root cause analysis showed that positive bacterial tests were related to inappropriate specimen handling, and bacteria identified were from skin microbiome. No patient had a positive blood culture post infusion, and no patient experienced any clinical or laboratory symptoms of bacteremia
- Typical steroid-related adverse events (Cushingoid features, hirsutism, central obesity, acne, skin atrophy, insomnia, and steroid-related behavioral problems) were not reported in EryDex-treated patients

	Placebo (N=19)	EryDex (N=116)
Pyrexia	16%	29%
Vomiting	16%	21%
Cough	32%	20%
Nasopharyngitis	26%	18%
Positive Bacterial Test*	21%	17%
Upper Respiratory Infection	5%	15%
Diarrhea	26%	12%
Pruritus	0	11%
Headache	11%	11%

*Positive bacterial test from the product not resulting in bacteremia in patients.

CONCLUSIONS

- Reported TEAEs were generally similar between EryDex- and placebo-treated patients with the exception of pruritus, a known complication occurring during intravenous infusion of dexamethasone phosphate
- Side effects typically attributed to chronic steroid use, such as Cushingoid features, hirsutism, or hypertension, were not observed
- The observed safety profile for patients with A-T treated in the ATTeST study suggests that EryDex may be a promising investigational agent for pediatric patients requiring chronic steroid use to control their disease

DISCLOSURES

This research was funded by EryDel and Quince Therapeutics. Biljana Horn, Dirk Thye, and Maureen Roden are employees of Quince Therapeutics.